Anti-PD-1 Therapy Reduces Bone Lesion Growth in a Novel Syngeneic Bladder Cancer Bone Metastasis Model

AACR 2018
Poster number 709

Tiina E. Kähkönen[1], Mari I. Suominen[1], Jussi M. Halleen[1], Jenni Bernoulli[1], Arne Scholz[2]
[1]Pharmatest Services, Turku, Finland; [2]Bayer AG, Berlin, Germany

E-mail correspondence to Tiina Kähkönen (tiina.kahkonen@pharmatest.com)

Introduction

Bladder cancer is a common and aggressive cancer occurring both in women and men. At diagnosis, lymph node involvement is frequently observed and in some cases metastatic spread can already be seen at early stage. With advanced stage, the metastatic incidence increases and the most common metastatic sites are bones, lungs, liver and peritoneum. Bone metastases count about 50% of all metastases and the 5-year survival rate in metastatic disease is only 15%. Bone metastases are incurable but can be treated to reduce tumor burden in bone, and new therapies such as immunotherapies hold the potential to treat patients with bone metastatic bladder cancer.

The aim of the study was to establish a novel syngeneic model for bladder cancer bone metastasis that could be used to study efficacy of new immunotherapies, and to test efficacy of anti-PD-1 therapy in the model.

Materials and Methods

The study consisted of two parts, validation and efficacy. In the model validation, 5x10^5 or 2x10^6 murine MBT-2 bladder cancer cells (Riken Cell Bank) were inoculated intratibially to 5-6 weeks old female C3H/HeN mice (Envigo, n=6). Tumor-induced bone changes (bone lesions) were followed by X-ray imaging once a week. At sacrifice, tumor-bearing tibias were collected and evaluated by histology. Immunohistochemical staining for PD-1 (Nordic Biosite) was performed for de-calcified tumor sections. In the efficacy study, 5x10^5 MBT-2 cells were inoculated intratibially. The mice were stratified to treatment groups (n=10 per group) based on similar bone lesion areas 10 days after cancer cell inoculation and treated with anti-PD-1 therapy (RMP1-14) or isotype control (rat IgG2a, both BioXcell 200 µg per dose) at Q3D schedule. Bone lesion growth was followed by X-ray imaging one week after start of treatment and at sacrifice.

Model validation

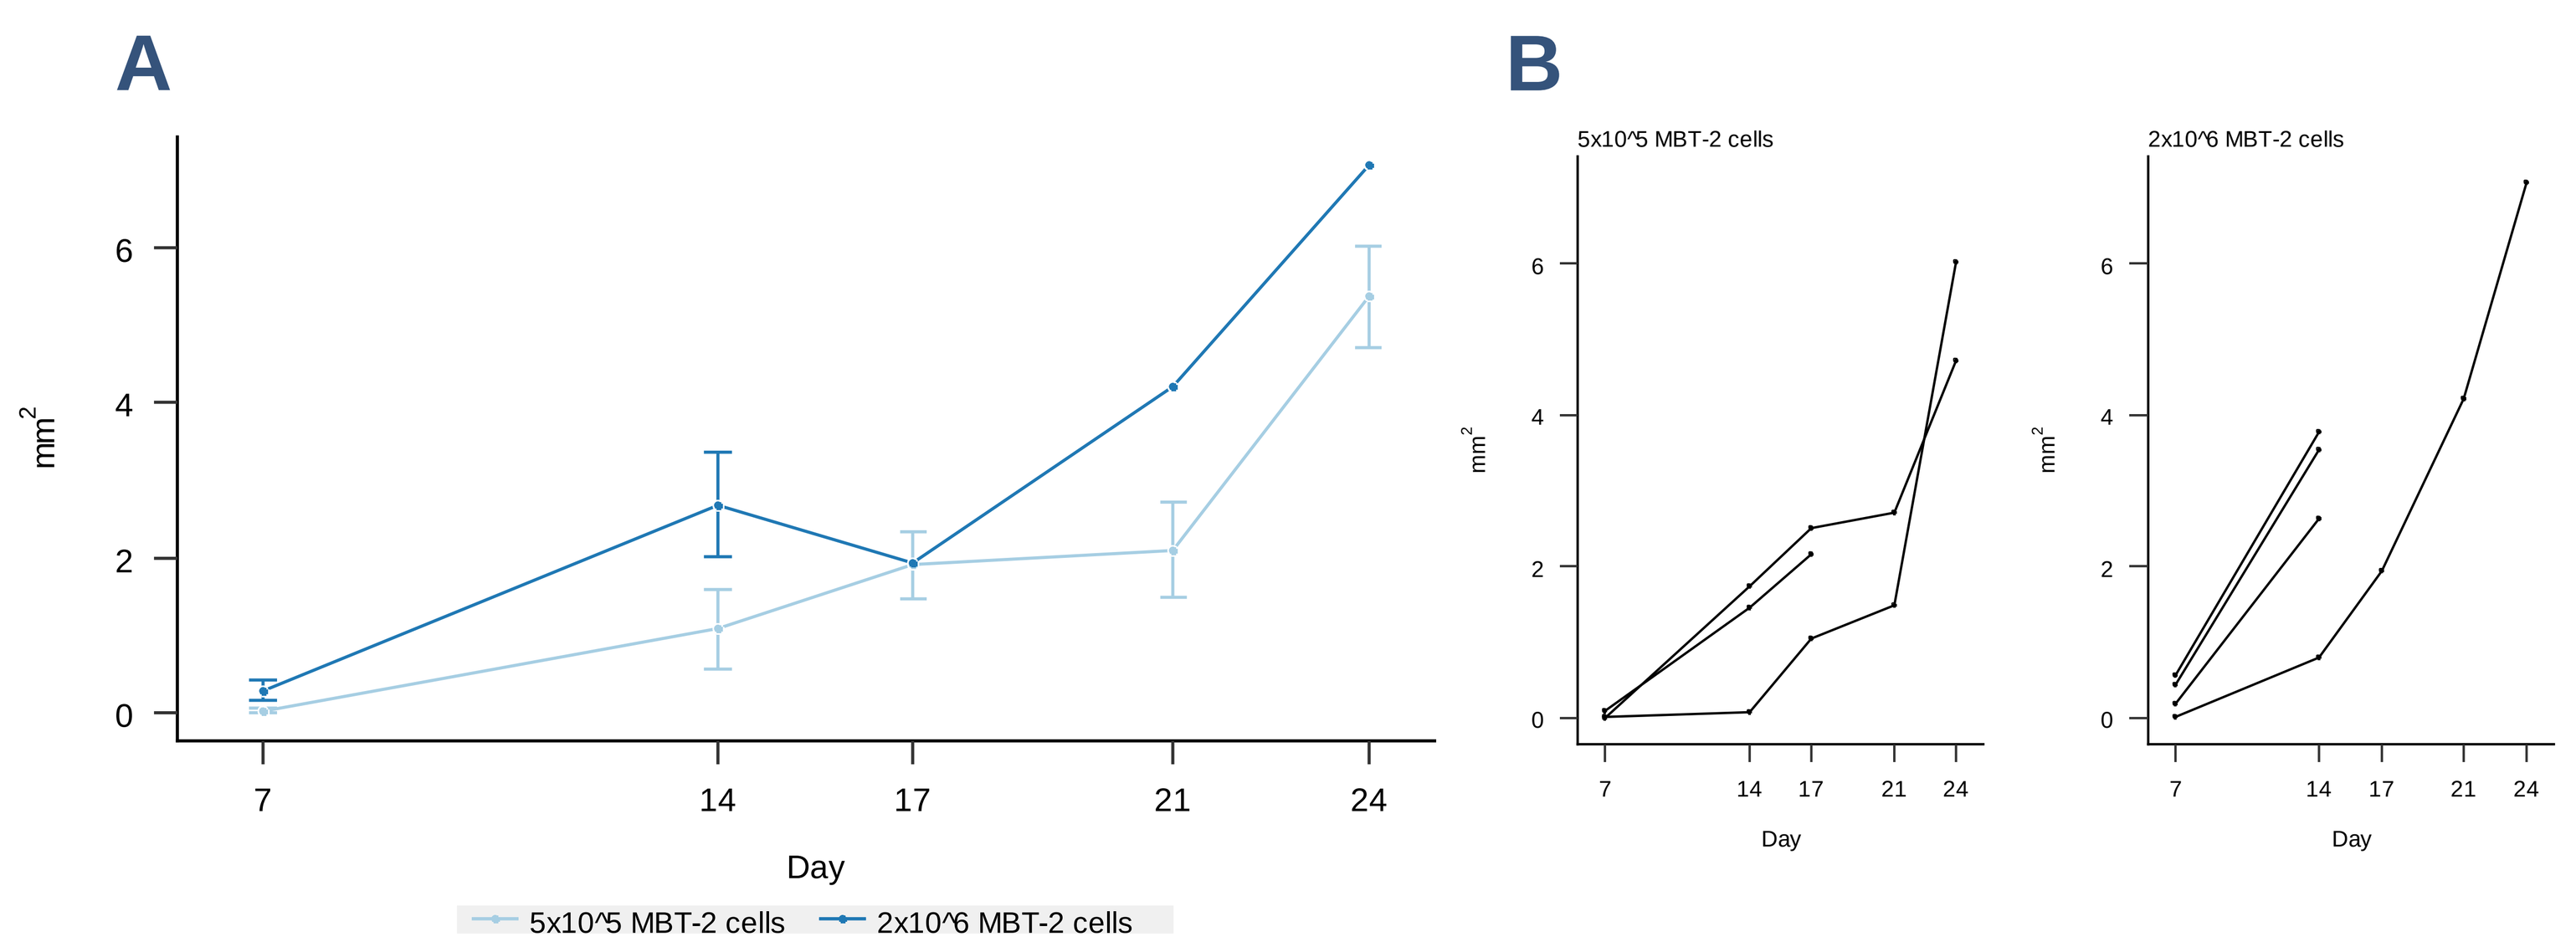
A
B
5x10^5 MBT-2 cells
2x10^6 MBT-2 cells
mm2
Day
5x10^5 MBT-2 cells
2x10^6 MBT-2 cells

FIGURE 1. A) Osteolytic bone lesion area (mm², mean ± SEM) and B) individual lesion growth charts are presented. The mice were sacrificed individually when extensive osteolytic lesions were observed (between study days 14 - 24).

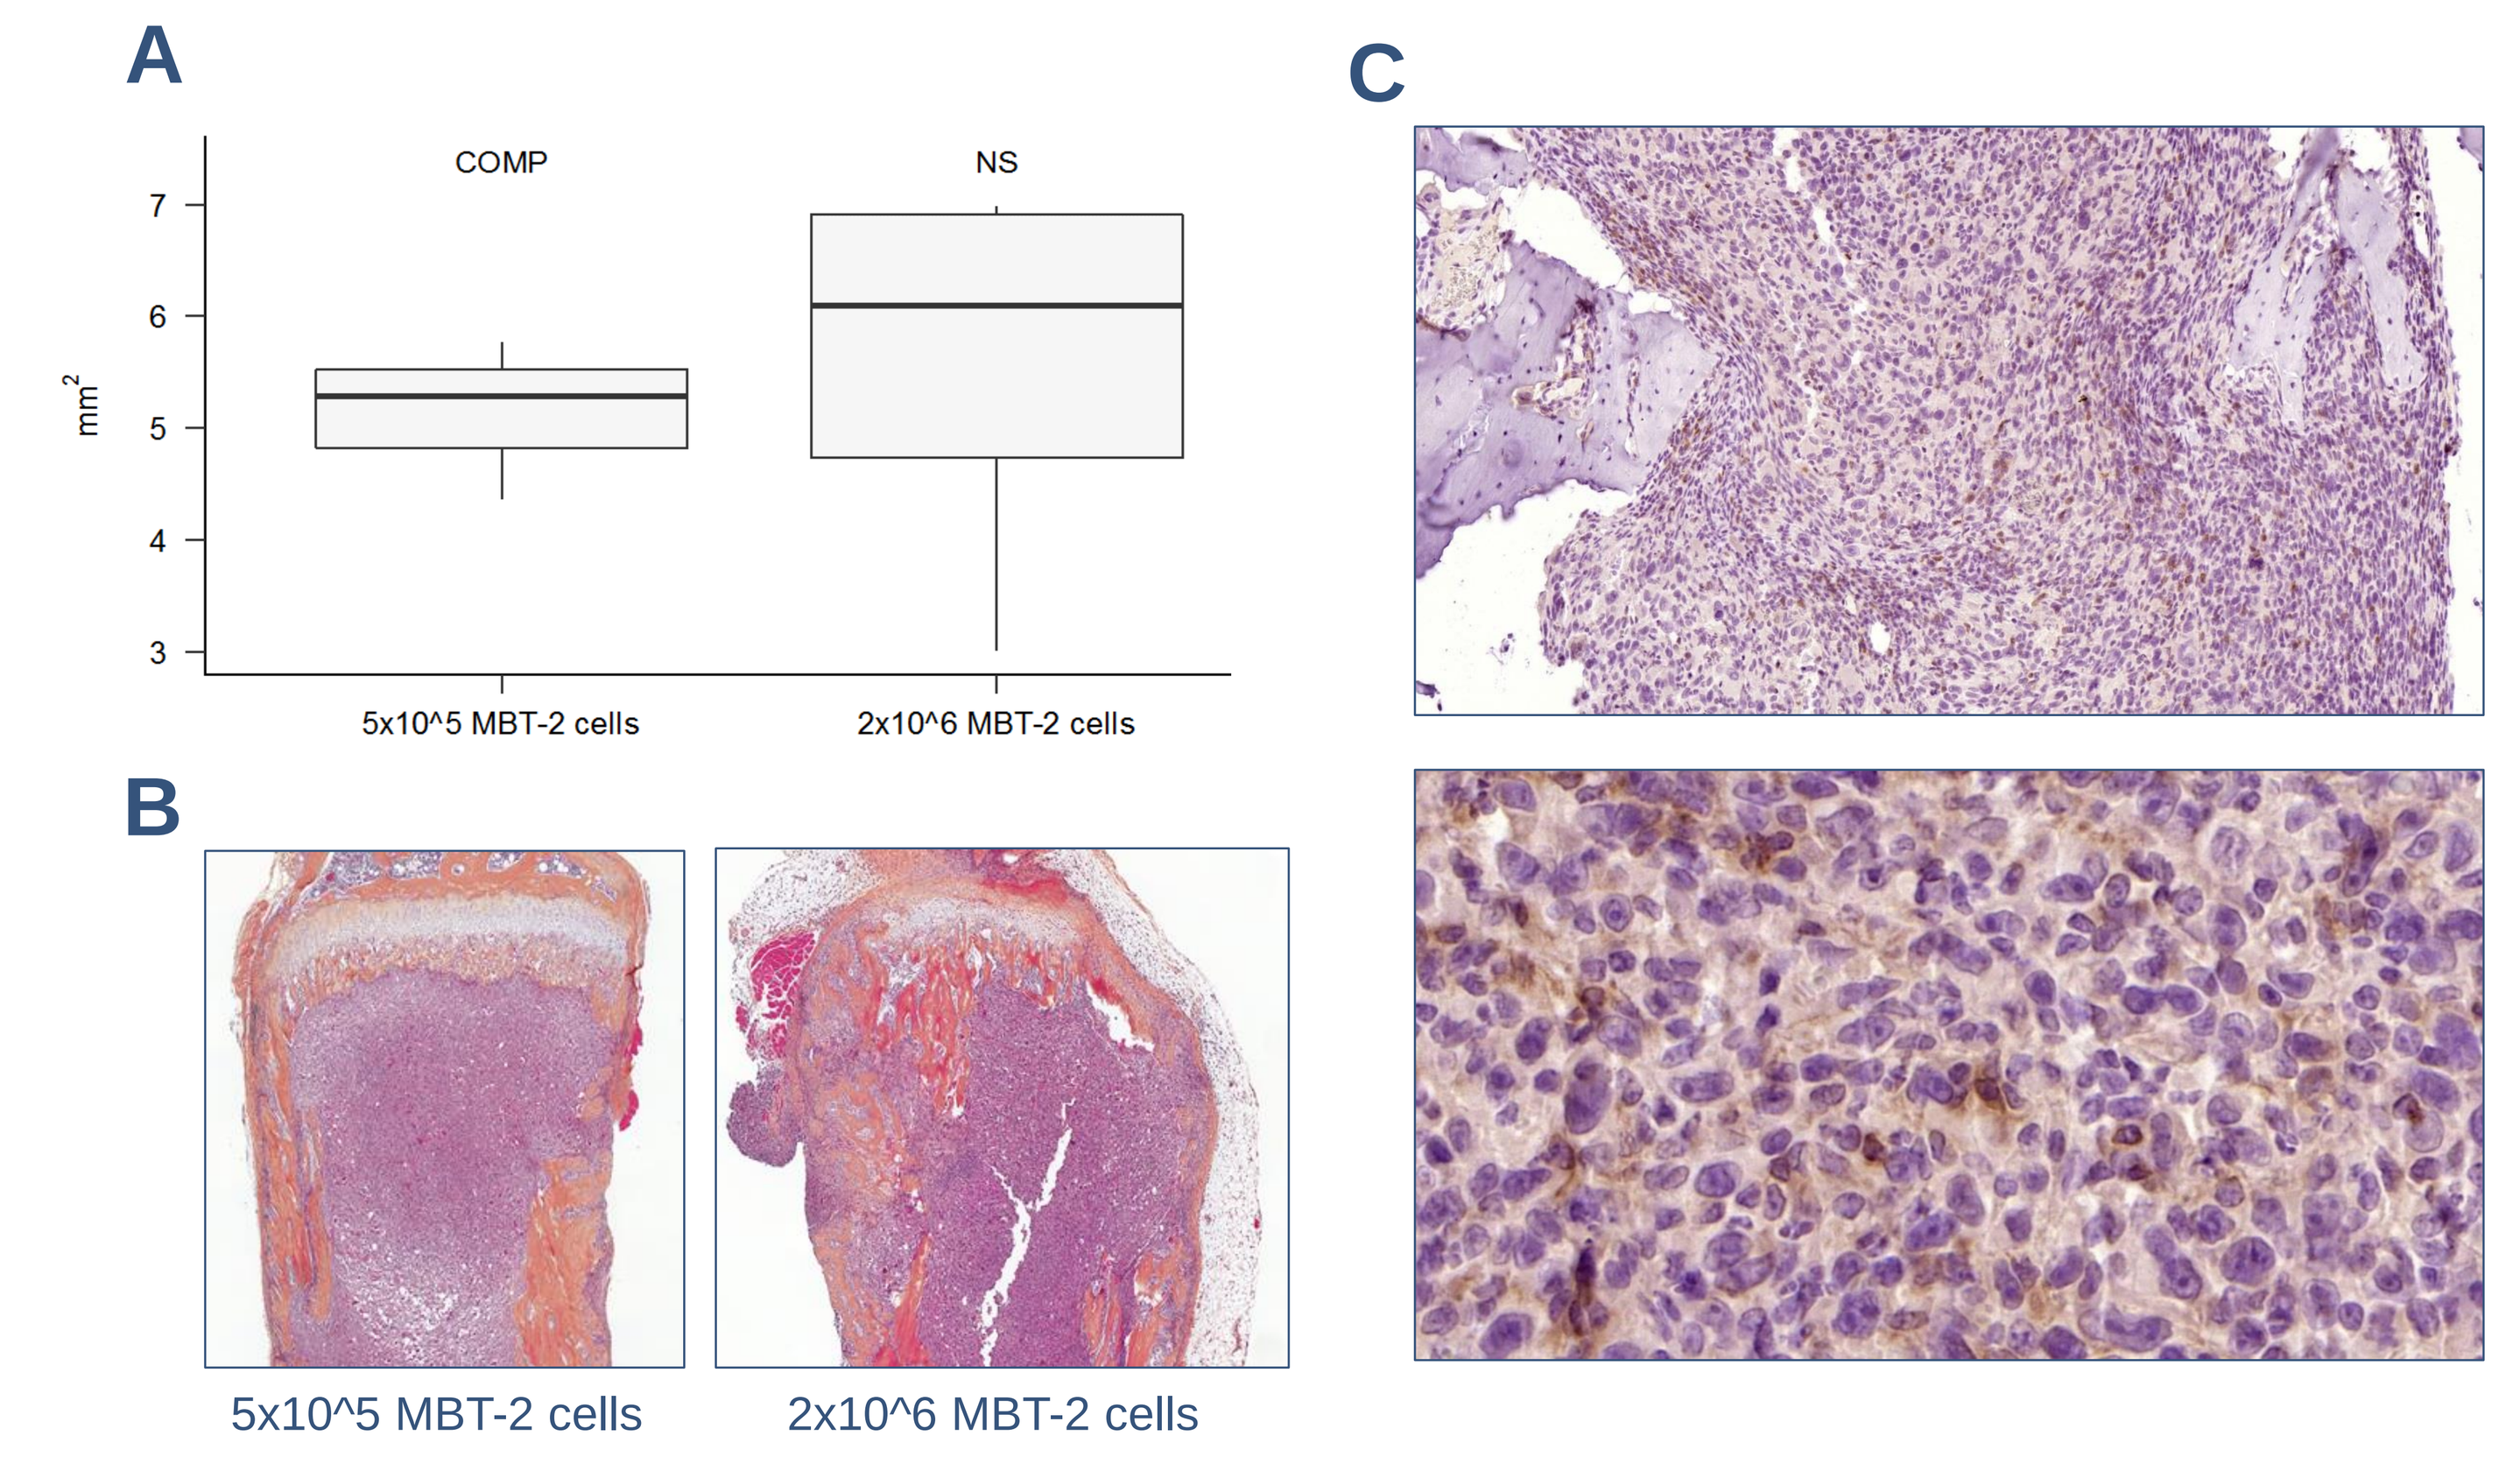
A
B
C
COMP
NS
mm2
5x10^5 MBT-2 cells
2x10^6 MBT-2 cells
5x10^5 MBT-2 cells
2x10^6 MBT-2 cells

FIGURE 2. A) Histological tumor area (mm², median ± IQR25% ± min/max) quantified from hematoxylin and eosin (HE) stained histological sections at endpoint. No statistically significant differences were observed between the study groups (p>0.05). Abbreviations: COMP = comparison group, NS = non-significant. B) Representative images from HE-stained tumor sections, magnification 4x. C) Representative images of PD-1 staining (magnification 20x in upper and 80x in lower image).

Efficacy study

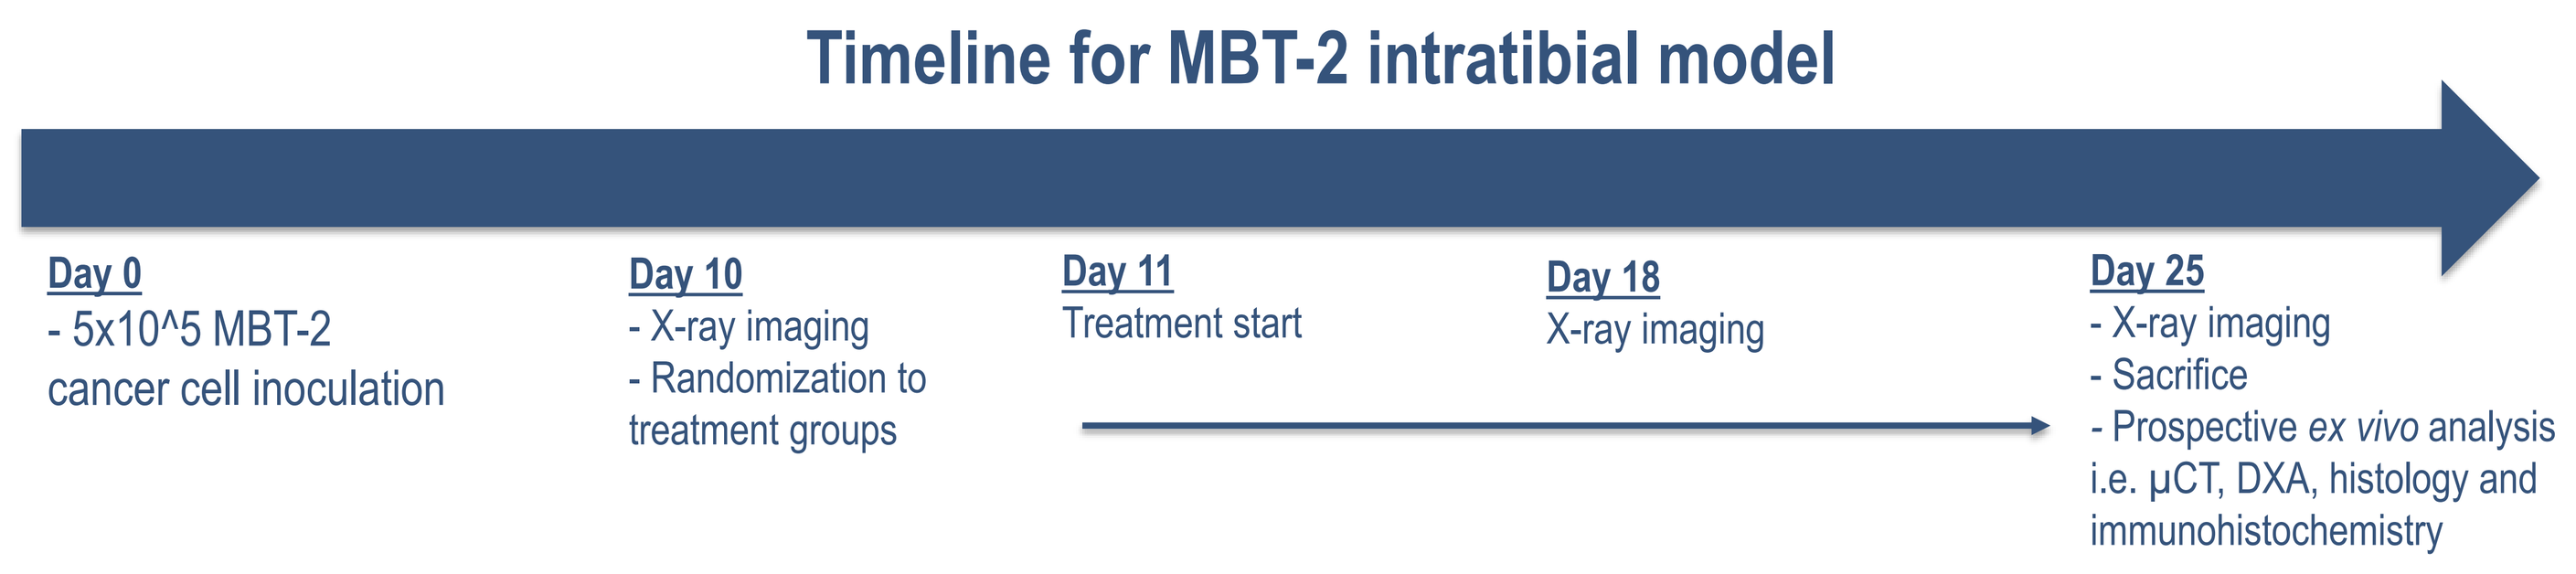
Timeline for MBT-2 intratibial model
Day 0
- 5x10^5 MBT-2 cancer cell inoculation
Day 10
- X-ray imaging
- Randomization to treatment groups
Day 11
Treatment start
Day 18
X-ray imaging
Day 25
- X-ray imaging
- Sacrifice
- Prospective ex vivo analysis i.e. µCT, DXA, histology and immunohistochemistry

FIGURE 3. MBT-2 bladder cancer cells were inoculated to proximal tibia of C3H mice at day 0. The mice were stratified to treatment groups at day 10 based on bone lesion area. Treatment was started at day 11. Treatment response was followed by X-ray imaging after one week on treatment (day 18) and at sacrifice (day 25).

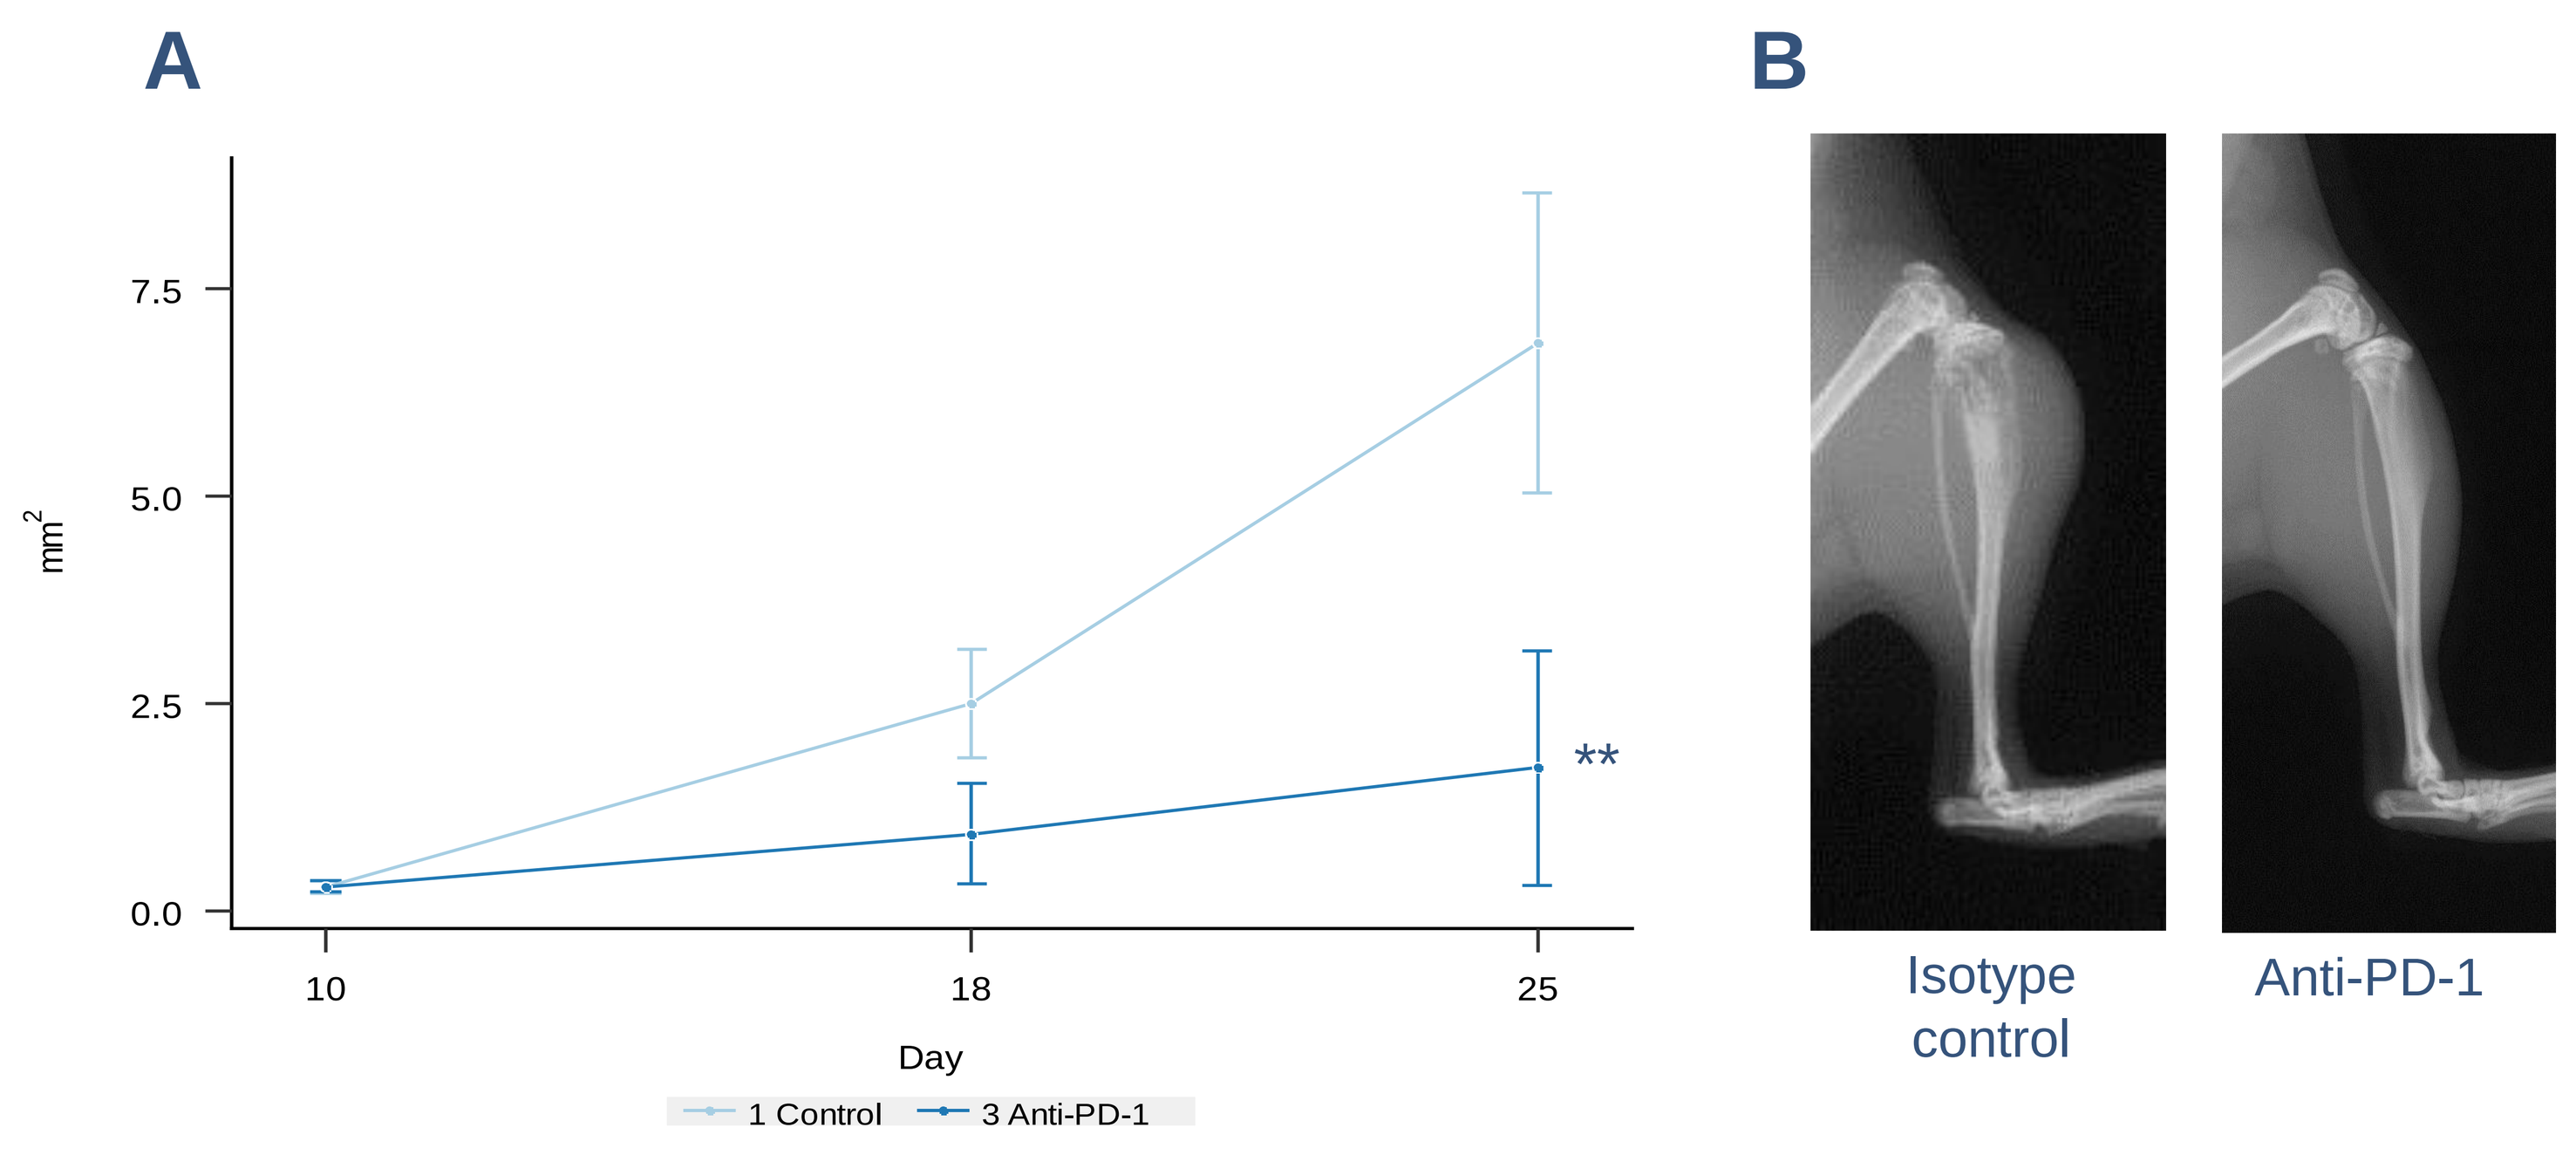
A
B
mm2
Day
**
1 Control
3 Anti-PD-1
Isotype control
Anti-PD-1

FIGURE 4. A) Quantitation of bone lesion area (mm², mean ± SEM) at randomization and one and two weeks on treatment. Anti-PD-1 treatment reduced bone lesion growth (p<0.01 **). B) Representative X-ray images from control and anti-PD-1 treated mice at endpoint.

Summary

- In the model validation study, a tumor take rate of 50 - 60% was observed based on X-ray imaging and histological findings at endpoint.
- MBT-2 cells induced an osteolytic bone reaction resulting in substantial bone loss in tumor-bearing tibia.
- The osteolytic bone lesions started to be visible between 7-14 days and the mice were sacrificed between 14-24 days due to extensive osteolytic lesions.
- Histology confirmed large tumors with PD-1 positive cells.
- In the efficacy study, treatment with anti-PD-1 reduced bone lesion growth.
- A trend towards decreased bone lesion area was seen already after one week of treatment, and a significant reduction of bone lesion area was observed at sacrifice.

Conclusions

A novel syngeneic bladder cancer bone metastasis model was established. Tumors were rapidly growing and induced osteolytic bone lesions typical for the patients with bladder cancer bone metastases.

Anti-PD-1 treatment reduced tumor induced bone changes in the model. Currently, novel drugs targeting immune checkpoints are under development and the model can be used when assessing efficacy of novel mono- and combination therapies against advanced bladder cancer.

Acknowledgements

BioSiteHisto Ltd is acknowledged for performing immunohistochemical stainings.

www.pharmatest.com